Inhibitors Of The CFTR Chloride Ion Channel As Potential Treatment For Acute Secretory Diarrhea: Development Of 6-membered Heterocycles

Doyle K[1], Peach J[1], Russell M[1], Penrose S[1], Van de Poël A[1], Jones G[1], Macritchie J[1], MacLeod A[1], Brückner S[1], Shearer J[1], Cronk D[1], Clark G[1], Beresford A[1], de Hostos E[2], Nguyen T[2], Brown D[2]

[1]BioFocus, Chesterford Research Park, Saffron Walden CB10 1XL, UK, email: info@glpg.com
[2]The Institute for OneWorld Health, 50 California St, Suite 500, San Francisco, CA 94111, USA

1. Introduction

Acute secretory diarrhea is responsible for the deaths of approximately 2 million children each year in the developing world.[1] The current standard of care is a glucose/salt solution, known as oral rehydration therapy (ORT), which is a palliative treatment for the disease. However, the labor intensive regimen alongside the lack of symptomatic relief leads to a high level of noncompliance. The identification of an adjunct therapy to work alongside ORT and improve time to symptomatic relief is paramount.

Acute secretory diarrhea is caused by an intestinal infection with pathogens that include *vibrio cholerae* resulting in the output of large volumes of watery stools. The major mechanism for this infection is the over-activation of the chloride ion channel, CFTR (cystic fibrosis transmembrane conductance regulator), present in the epithelial cells within the intestinal wall.

Small molecule CFTR inhibitors have been reported to be efficacious in *in vivo* models of secretory diarrhea.[2,3] The aim of this work was to develop novel and effective CFTR inhibitors suitable for clinical evaluation in the treatment of secretory diarrhea.

2. Literature inhibitors

GlyH-101[2]
- Acyl hydrazone. Contains toxic β-naphthylamine moiety
- Cell assay (FRT-YFP) IC_{50} ~5 μM
- Active in closed-loop murine model
- Site of action reported to be 'extra-cellular'

$CFTR_{inh}$-172[3]
- 2-Thioxo-thiazolidin-4-one series
- Cell assay (FRT-YFP) IC_{50} 300 nM
- Active in closed-loop murine model
- Site of action reported to be 'intra-cellular'

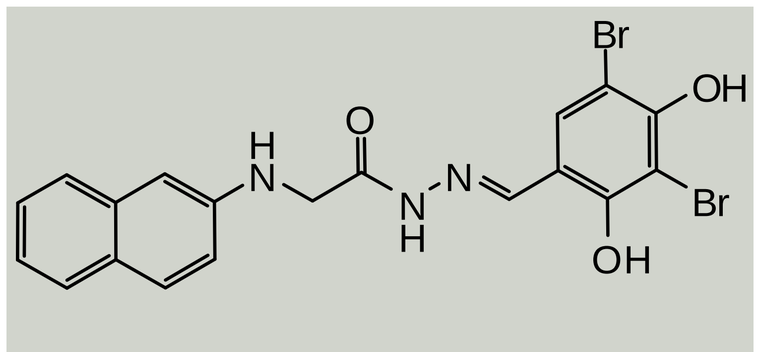

GlyH-101

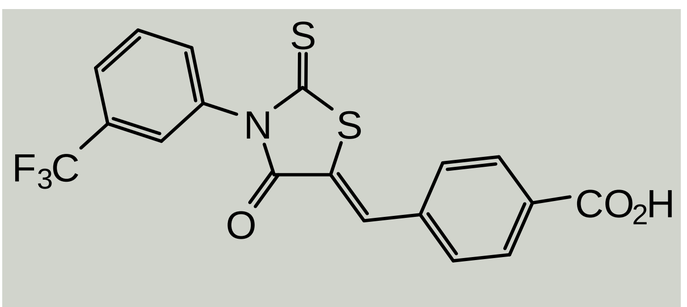

$CFTR_{inh}$-172

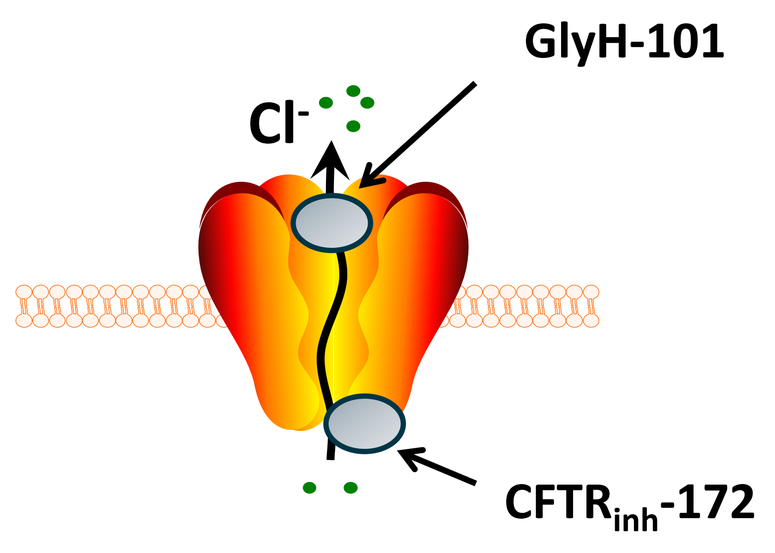
Cl-
GlyH-101
CFTRinh-172

3. Assays utilized

In vitro cell-based assays

- T84 Cell Line (primary assay)
 - Human colonic epithelial cell line, endogenously expresses CFTR
 - Suitable for FLIPR assay, using membrane-potential sensitive dye
- CHO-CFTR Cell Line (secondary assay)
 - Generated CHO cell line that heterologously expresses hCFTR
 - Inducible expression factor required
 - Suitable for use in FLIPR, using membrane-potential sensitive dye
 - Suitable for use in IonWorks® Quattro™ (IWQ) electrophysiology assay

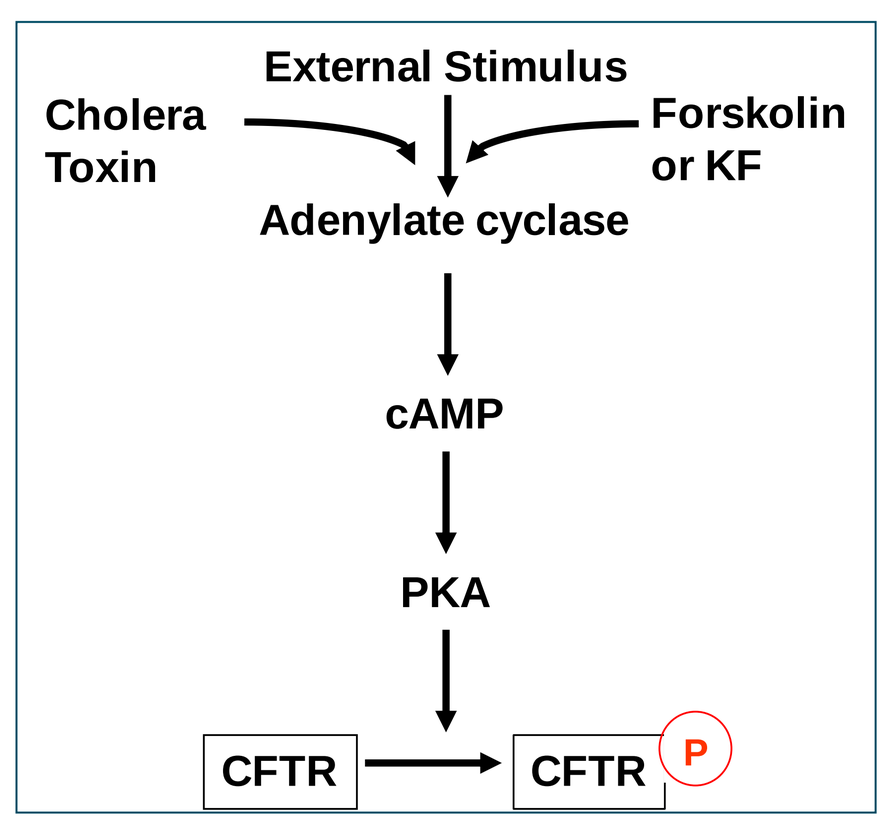
External Stimulus
Cholera Toxin
Forskolin or KF
Adenylate cyclase
cAMP
PKA
CFTR
CFTR
P

In vivo animal models

Two surgical models both measuring cholera toxin (CTX) induced secretion

Species	Surgery	Route of administration	End-point
Mouse	Intestinal loop: 2 loops per mouse (1 Cholera toxin & 1 PBS, +/- compound)	*intra-lumenal* with CTX	Secretion measured as loop g/cm. Reported as %reduction in loop swelling compared to control loop
Rat	Cecectomized	*po*	Fecal Output Index: Sum of number of defecation episodes and fecal consistency score for each episode

4. Series evolution

Medicinal chemistry efforts on the acyl hydrazone series led to the discovery of proprietary acyl hydrazones that retained cellular potency.[4]

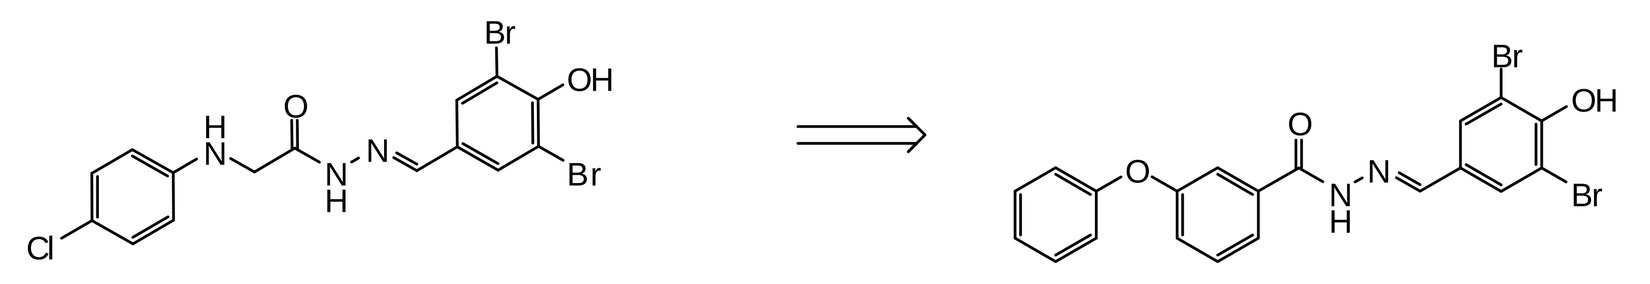

GlyH-114
T84 IC_{50} 9.6μM

T84 IC_{50} 6.8μM

Conformational analysis of acyl hydrazones indicated two energetically favored conformers: syn-*trans* and anti-*cis*. More than 20 heterocyclic systems were synthesised leading to a wide selection of active leads. Some of these leads evolved into a distinct series of 6-membered heterocyclic compounds, as exemplified by the 1,2,4-trazine motif. The newly discovered heterocyclic analogues were found to be efficacious in an *in vivo* model for secretory diarrhea.

conformer	$\Delta E_{HF/6\text{-}31G}$ (kcal/mol)
Syn *trans*	**0**
Syn *cis*	31.56
Anti *trans*	24.47
Anti *cis*	**3.14**

Table: *Ab initio* calculations carried out on simplified acylhydrazone

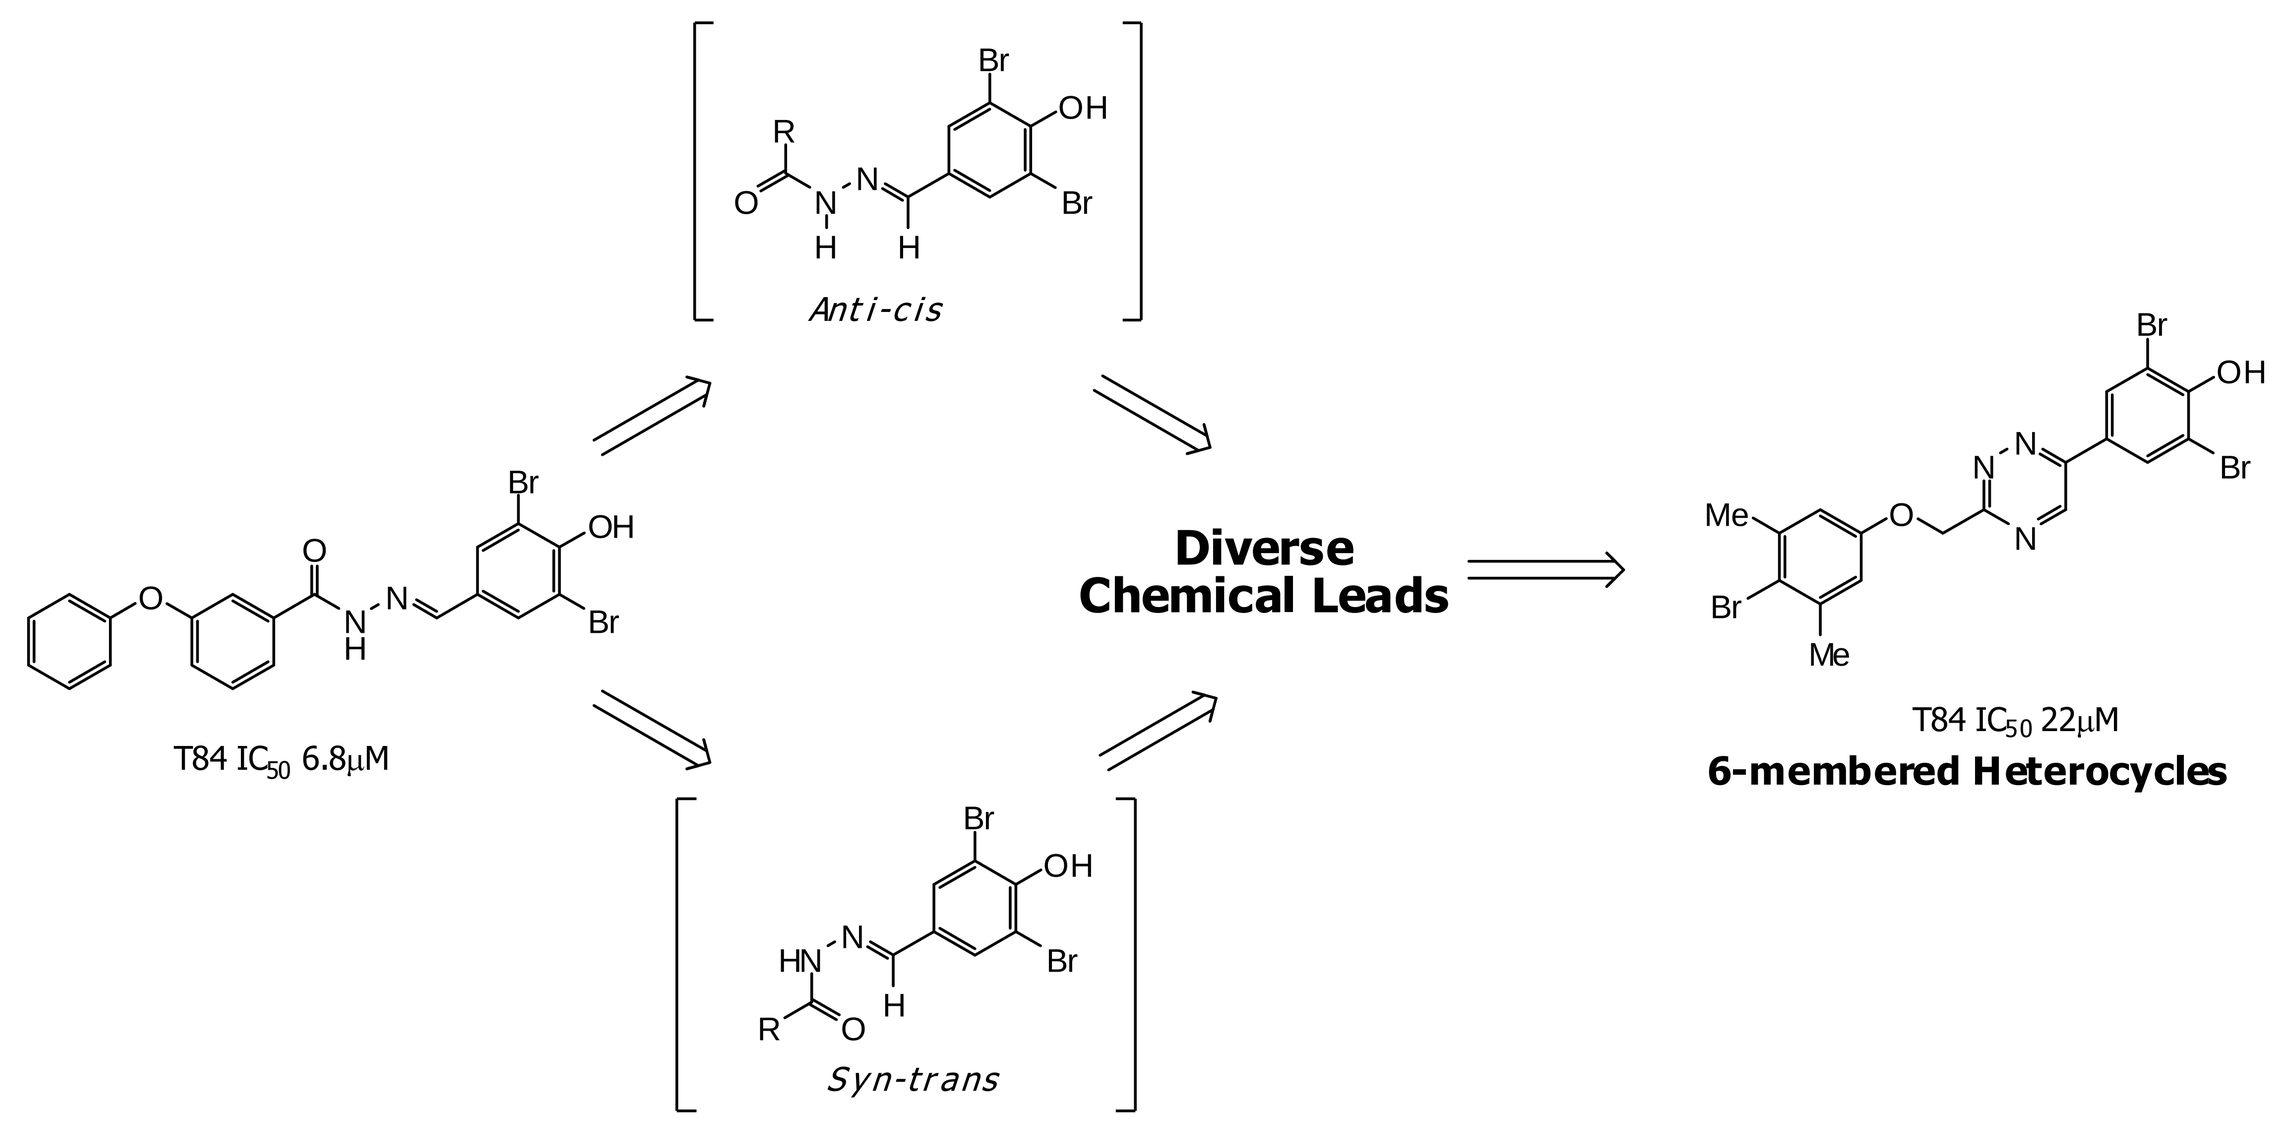
Anti-cis
Syn-trans
T84 IC50 6.8μM
Diverse Chemical Leads
T84 IC50 22μM
6-membered Heterocycles

5. 1,2,4-triazine series

Acyl hydrazone bioisosteres

A selection of analogous acyl hydrazones and 1,2,4-triazines were compared side-by-side, three examples of which are shown (see **Table**). Comparison of T84 FLIPR data showed the 1,2,4-triazine core to be bioisosteric with acyl hydrazones. Based on this observation, the 1,2,4-triazine system was explored further.

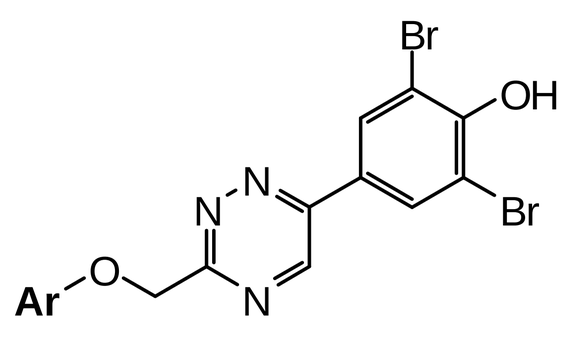

1,2,4-Triazine

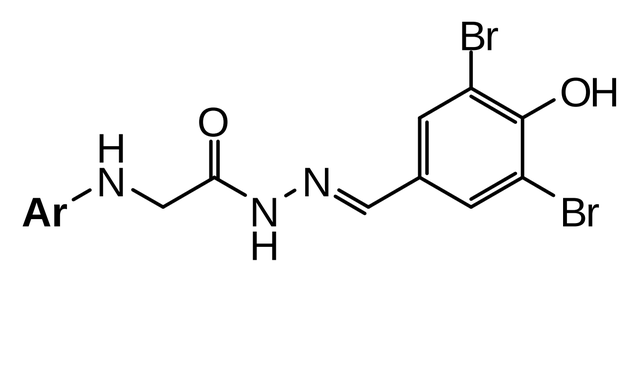

Acyl hydrazone

T84 IC_{50} (μM)	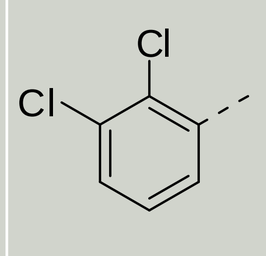	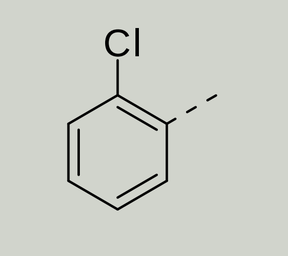	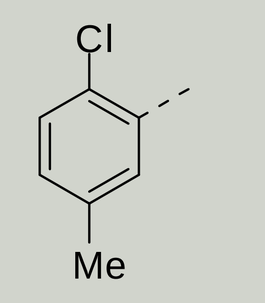
1,2,4-Triazine	8.6	>30	6.4
Acyl hydrazone	10.2	-6% @ 50μM	17.8

Table: Comparison of acyl hydrazone and 1,2,4-triazine analogues

6. 1,2,4-triazine series

Preparation of 1,2,4-triazines

The initial preparation of 1,2,4-triazines was achieved in one pot using commercially available acyl hydrazides. Although the route allowed rapid access to the target compounds, it often gave a complex reaction mixture, worked best with oxyacetic acid hydrazides and so had a limited scope for expansion.

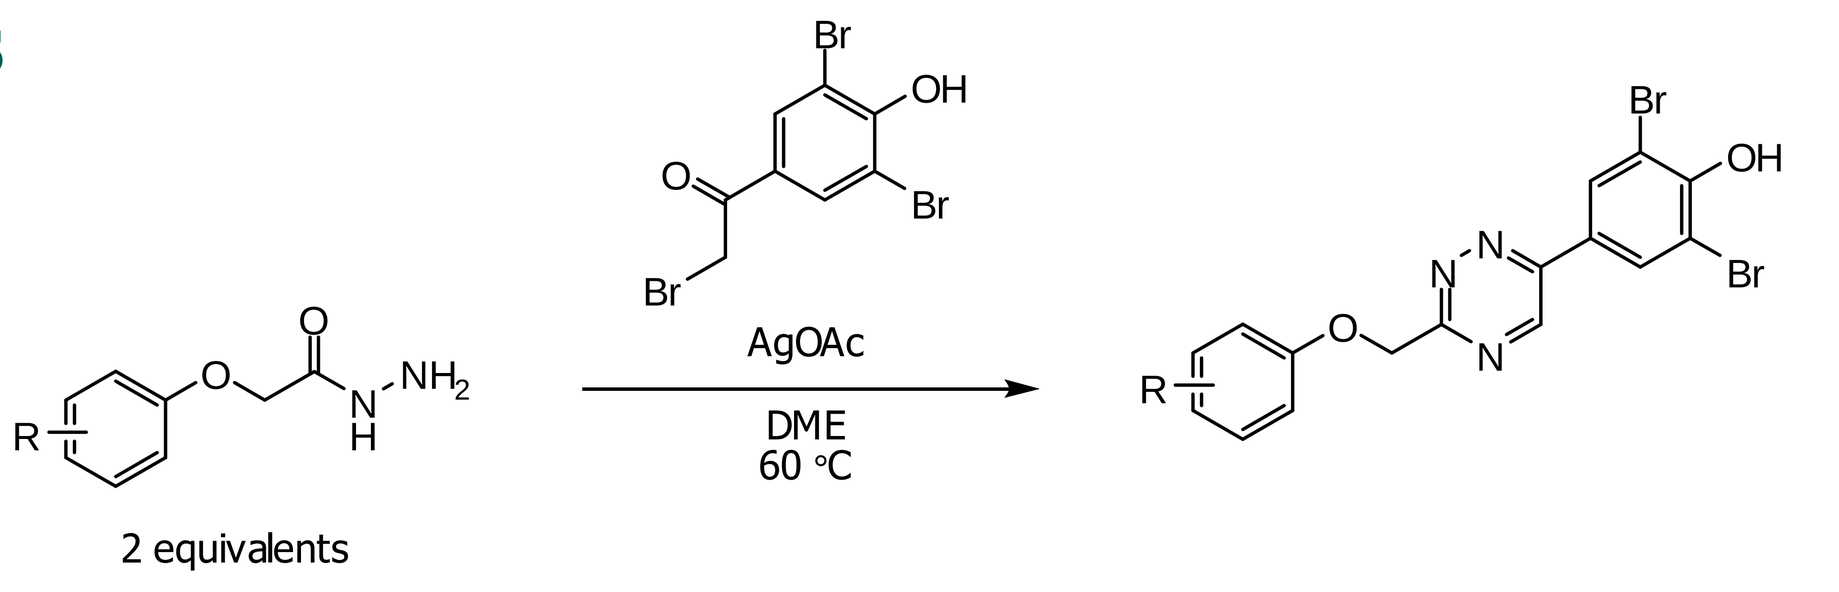
2 equivalents
AgOAc
DME
60 °C

Scheme: Initial preparation of 1,2,4-triazines

Reversing the methyleneoxy linker allowed activity to be retained and lead to greater chemical diversity.

Reversing the linker

The key building block (2,6-dibromo-4-(3-methylsulfanyl-[1,2,4]triazin-6-yl)-phenol), was prepared. The methylsulfanyl group was subsequently displaced with a range of alcohols, allowing rapid exploration of the series.

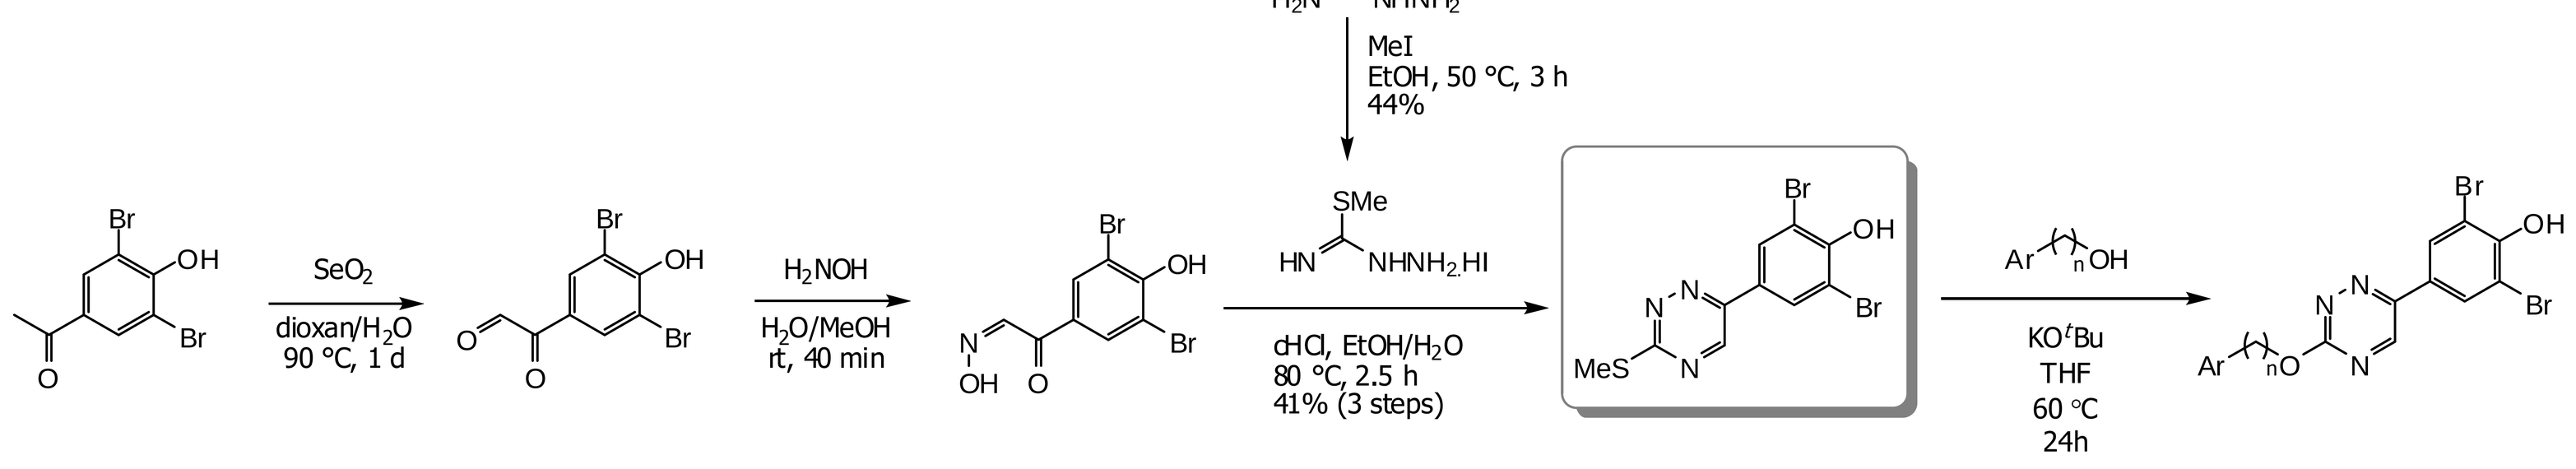
MeI
EtOH, 50 °C, 3 h
44%
SeO2
dioxan/H2O
90 °C, 1 d
H2NOH
H2O/MeOH
rt, 40 min
cHCl, EtOH/H2O
80 °C, 2.5 h
41% (3 steps)
KOtBu
THF
60 °C
24h

Scheme: Synthetic route to key building block, 2,6-dibromo-4-(3-methylsulfanyl-[1,2,4]triazin-6-yl)-phenol

On reversing the methyleneoxy linker it was observed that cellular potency was not only retained but often improved.

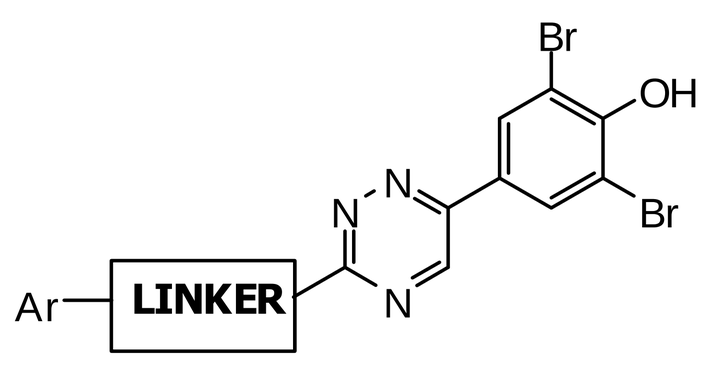

T84 IC_{50} (μM)	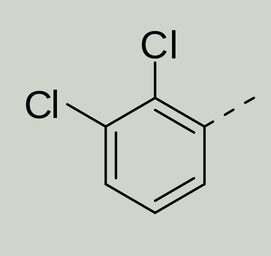	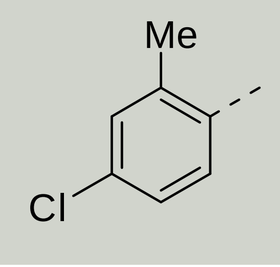	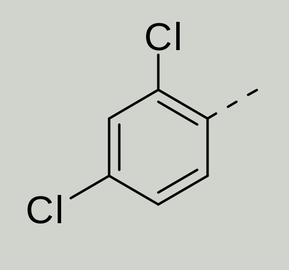	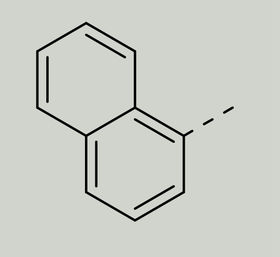
$-OCH_2-$	8.65	10.8	10.9	5.38
$-CH_2O-$	2.70	1.34	0.94	0.88

Table: Reversal of methyleneoxy linker

Linker expansion

Increasing the length of the *O*-linker group, cellular potency was maintained. However, this was at the detriment of physicochemical properties notably solubility (data not shown). Based on these observations, the methyleneoxy chain length was seen as optimum, balancing *in vitro* potency with suitable physicochemical properties.

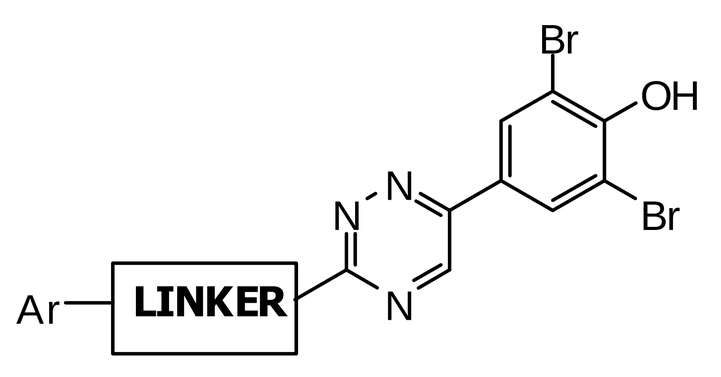

T84 IC_{50} (μM)	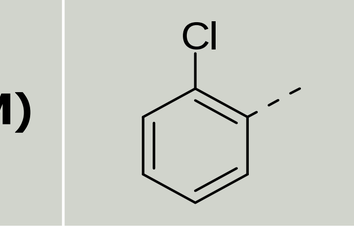	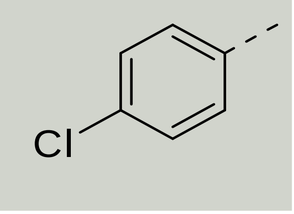	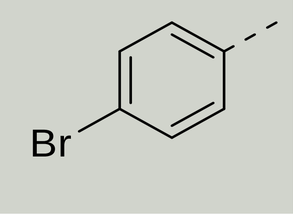	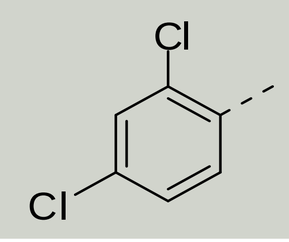	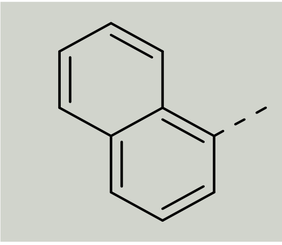	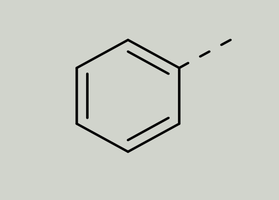
$-CH_2O-$	1.2	0.56	3.0	0.94	0.88	1.4
$-CH_2CH_2O-$	0.34	0.17	0.32	0.66	0.26	0.47
$-CH_2CH_2CH_2O-$	-	0.61	-	-	-	-
$-CH(CH_3)O-$	-	0.84	-	-	-	-

Table: Expansion of linker length

7. 1,2,4-triazine series: *In vivo* actives

In vivo profiling

Based on their cellular potencies and physicochemical profiles, a selection of 1,2,4-triazines were tested in the murine closed-loop model. The compounds were shown to be efficacious and inhibited cholera toxin induced secretion.

	Cell assays IC_{50} (μM)			Kinetic solubility @ pH7.4 (μM)	Caco-2		Murine closed-loop model (% inhibition)	
	T84 FLIPR	CHO-CFTR FLIPR	CHO-CFTR IWQ @ -60mV		A2B ($x10^{-6}$ cm/s)	Efflux	10 μg dose	100 μg dose
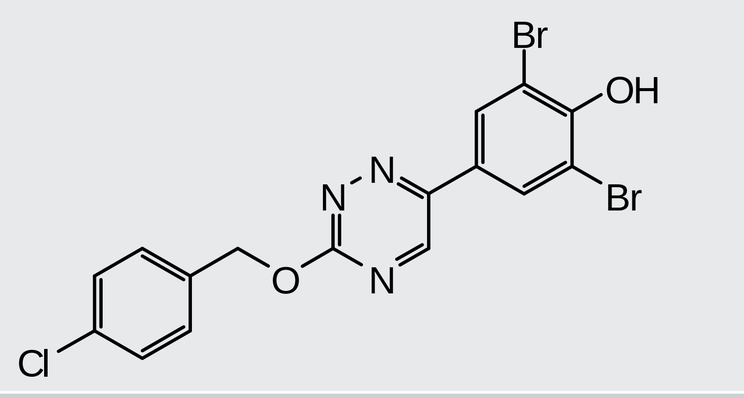	0.56	3.17	2.58	62	40.6	0.51	45	87***
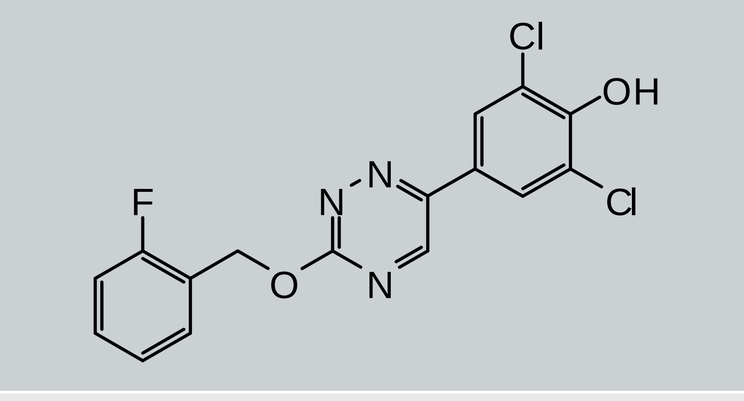	5.45	3.41	4.74	34	19.5	0.83	62*	70**
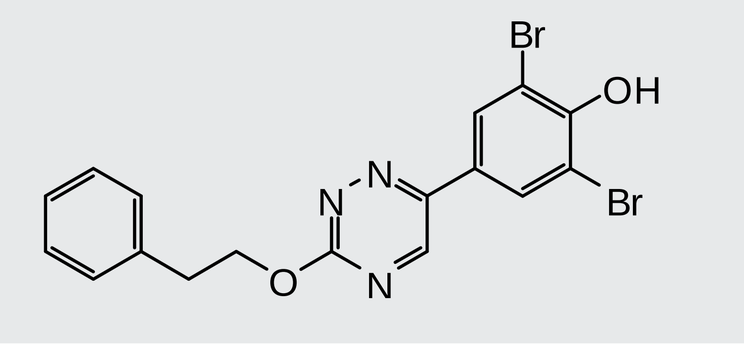	0.46	1.68	n/a	12	n/a	n/a	4	78**

Table: Selected profile data for 1,2,4-trazine compounds. Statistical significance is shown for the closed-loop model compared to the control group

8. Conclusions

- Conformational analysis of proprietary acyl hydrazone inhibitors, followed by a parallel synthesis approach, led to the discovery of novel heterocyclic CFTR inhibitors
- Analysis showed proprietary 1,2,4-triazine system to be bioisosteric with acyl hydrazones
- Originally discovered 1,2,4-triazine series was transformed into a more tractable 1,2,4-triazine series by preparation & exploitation of a key building block
- Optimization led to a series of 1,2,4-triazines that demonstrated statistical significant *in vivo* inhibition in a murine model of secretory diarrhea

9. References

1. Bryce, J. *et. al. Lancet* **2005**, *365*, 1147-1152
2. Verkman, A. *et. al. J. Gen. Physiol.*, **2004**, *124*, 125-157
3. Verkman, A. *et. al. J. Clin. Invest.*, **2002**, *110*, 1651–1658
4. Russell, MGN *et. al.* WO 08/121877

***IonWorks® Quattro™** is a trademark of Molecular Devices Corporation*

©Copyright 2010 Galapagos NV

Institute for OneWorld Health
A Nonprofit Pharmaceutical Company

BioFocus
A Galápagos Company